Randomized Controlled Trial in a Real-World Setting Shows a Light Touch "Food Is Medicine" Program Improves Health Behaviors and Self-Reported Health Status in People with Diabetes

Maggie Wilkin[1], MPH, Betty Sun[1], MS, RD,
Suki McClatchey[2], MPH

[1]Public Health Institute Center for Wellness and Nutrition, [2]Abbott Fund

This study and Abbott's Healthy Food Rx program were funded by Abbott Fund, the philanthropic foundation of the global healthcare company Abbott. Healthy Food Rx is part of Abbott and Abbott Fund's Future Well™ Communities initiative in Stockton, California.

BACKGROUND

Resources that enhance quality of life can have a significant influence on population health outcomes, including the provision of healthy food, high quality healthcare, transportation services, and access to education [1]. In San Joaquin County, California, where this study took place, there is a 13.9% prevalence of diabetes [2] and an estimated 48% of residents are at risk for developing diabetes. There is evidence that healthy food prescription programs can improve food security and diet quality providing benefit to people with chronic conditions, such as diabetes [3]. However, there are few comparison studies.

OBJECTIVE

- The objective of this randomized controlled trial (RCT) was to assess the impact of a low-touch Food is Medicine program, Healthy Food Rx, for people with diabetes in a highly food-insecure community compared to usual care.
- The results add to the growing body of evidence for Food is Medicine to improve well-being and quality of life for people with diabetes.

METHODS

- This RCT used a delayed intervention, randomized design to assess the impact of Healthy Food Rx on patients with diabetes at Community Medical Centers (CMC), a Stockton, CA federally-qualified health center (FQHC).
- Participants were randomized to Intervention or Control who received a delayed intervention, stratified by a baseline A1C value (A1C <9% versus 9% or higher). CMC staff were blind to the randomization, and all participants received usual care as well as social needs screening and referrals.
- Bilingual English/Spanish health educators were trained to conduct program outreach and enrollment, obtain research study consent, and administer the baseline and post-program surveys.
- Hemoglobin A1Cs were obtained from medical records. Post program A1Cs were considered valid if taken within 60 days before or 90 days after their last food box.

The Center for Wellness and Nutrition is a program of the Public Health Institute. For more information, contact Maggie Wilkin, Director of Research and Evaluation, margaret.wilkin@wellness.phi.org.

For more information about Healthy Food Rx, scan here.

HEALTHY FOOD RX PROGRAM

- 12 home-delivered food boxes, ~$40 value, over 6 months
- Boxes contained family-sized, recipe-based meal kit plus pantry staples, and local and seasonal produce
- Each box included ~8 cups of fruit and 12 cups of vegetables
- Nutrition education provided in the form of ~3 healthy text messages sent out per box and cooking videos
- Optional participation in diabetes education and social needs referrals (emergency food, non-medical transportation, community diabetes care clinics)
- Food sourcing, recipes, home-delivery, nutrition education provided by Emergency Food Bank of Stockton, CA.

Figure 1: Study Flow

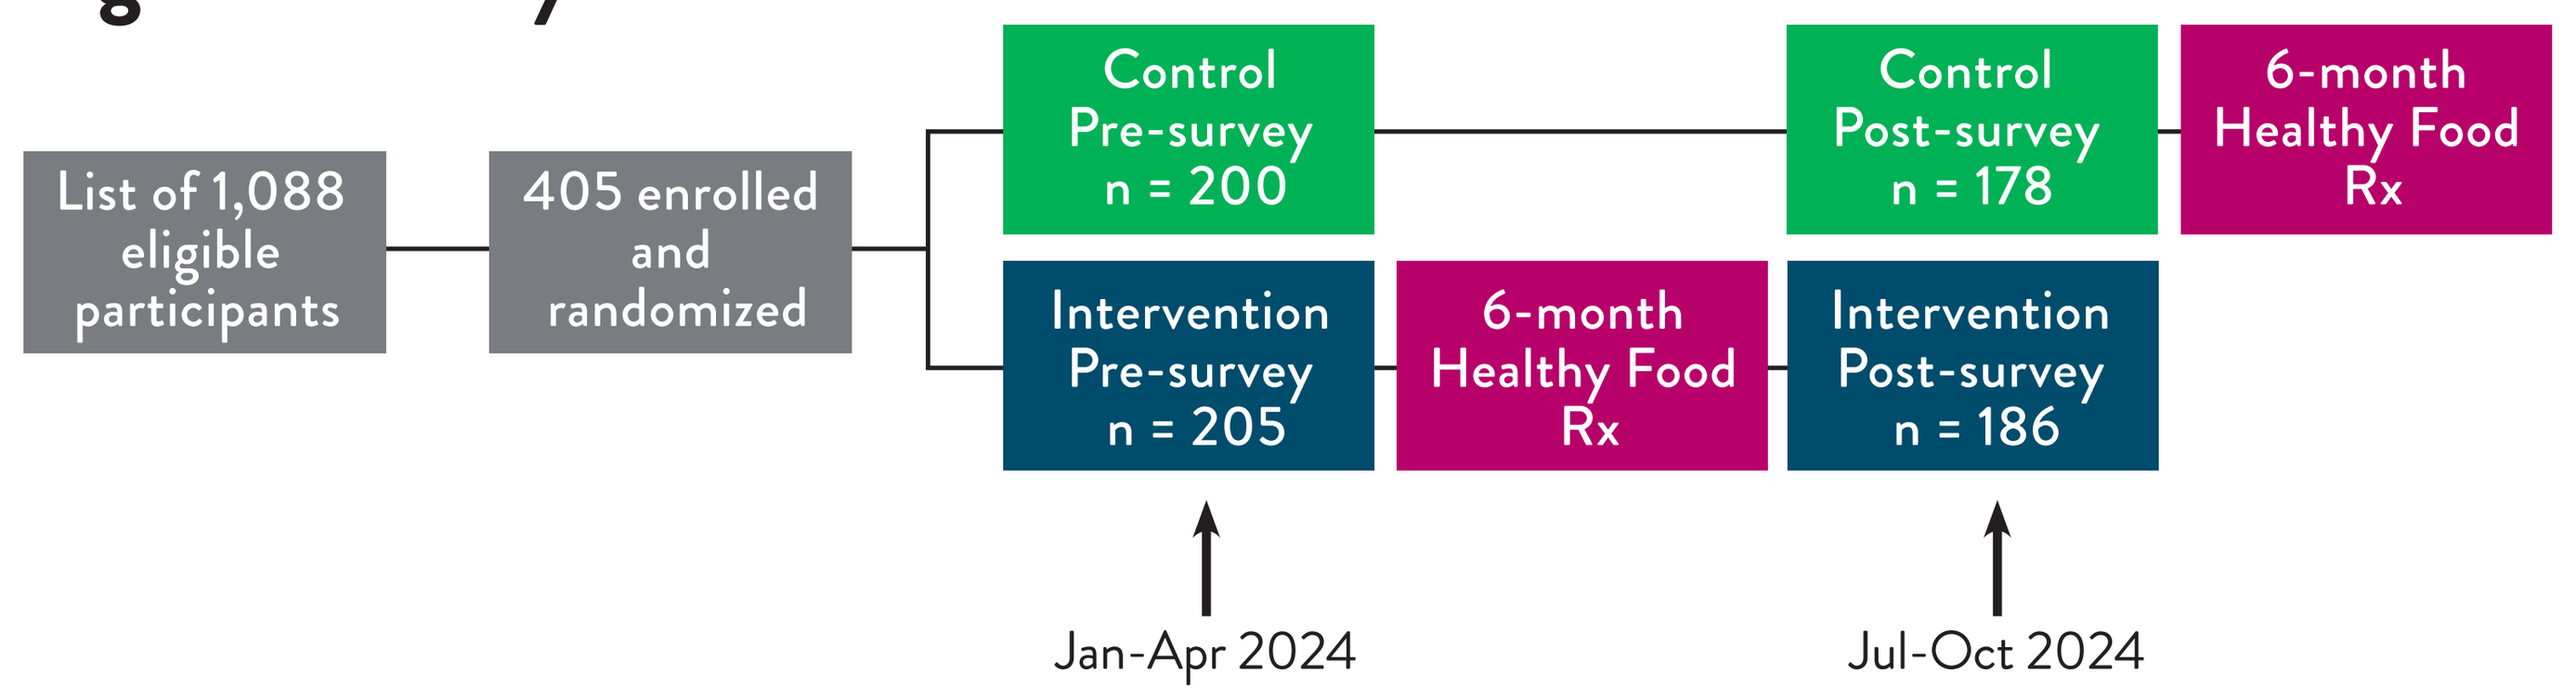
List of 1,088 eligible participants
405 enrolled and randomized
Control Pre-survey n = 200
Control Post-survey n = 178
6-month Healthy Food Rx
Intervention Pre-survey n = 205
6-month Healthy Food Rx
Intervention Post-survey n = 186
Jan-Apr 2024
Jul-Oct 2024

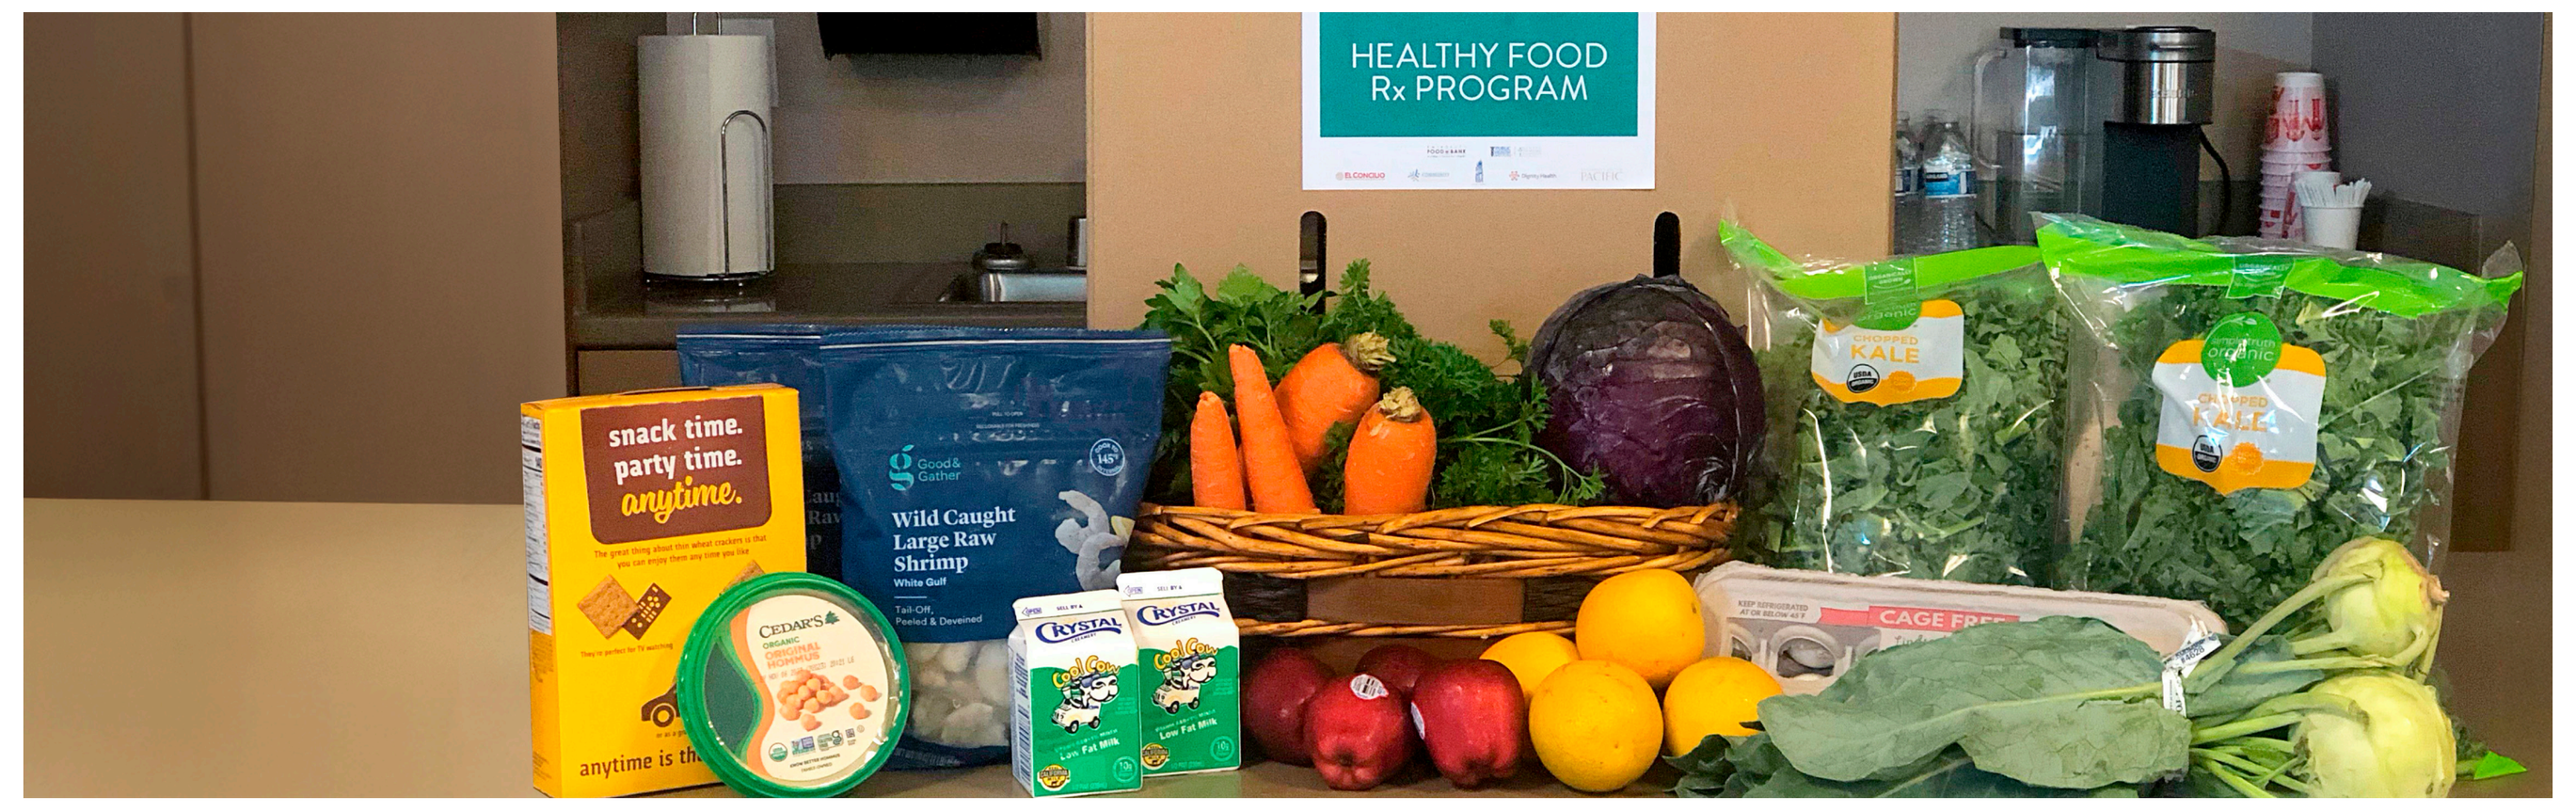
HEALTHY FOOD Rx PROGRAM
snack time. party time. anytime.
Wild Caught Large Raw Shrimp
CAGE FREE

LIMITATIONS

- Clinical information was not available, such as years with diabetes, number of visits, medications, etc., therefore it was not possible to control for those factors.
- All participants received comprehensive diabetes clinical care as part of usual care, which may have included medication changes and referrals to diabetes self-management programs, impacting outcomes for the control group as well as the intervention group.
- A1C values were gathered on the patient's normal lab schedule, which may not have captured the program's impact on glycemic control, however all measures were within 90 days of the end of the program.

"The program impacted me in different ways. I never had this kind of help before. The food box was delivered to my home. I did not have to go pick it up anywhere. It made me feel I was important too, que soy de la familia, gracias CMC. [that I am part of a family, thank you CMC.] The food made me stay healthier with the different recipes. I needed this food. Thank you very much."
– Healthy Food Rx participant

RESULTS

- The Intervention and Control groups were not significantly different at baseline: 60% were female, 86% identified as Hispanic or Latino, and 76% were over the age of 50.
- Both groups had statistically significant improvements in several outcomes, including A1C, water and fruit consumption.
- Intervention group participants improved significantly more than Control in vegetable consumption and self-reported health status.
- Food security was low in this population. The Intervention group improved slightly, although not significantly, after participation while Control declined.
- 98% of Intervention group participants were satisfied with the program and 99% indicated they were likely to recommend the program to others.
- 97% of participants used all or most of the food, and 67% shared the food with two or more people.

Figure 2. Mean A1C (%), by Group, Pre- and Post-survey

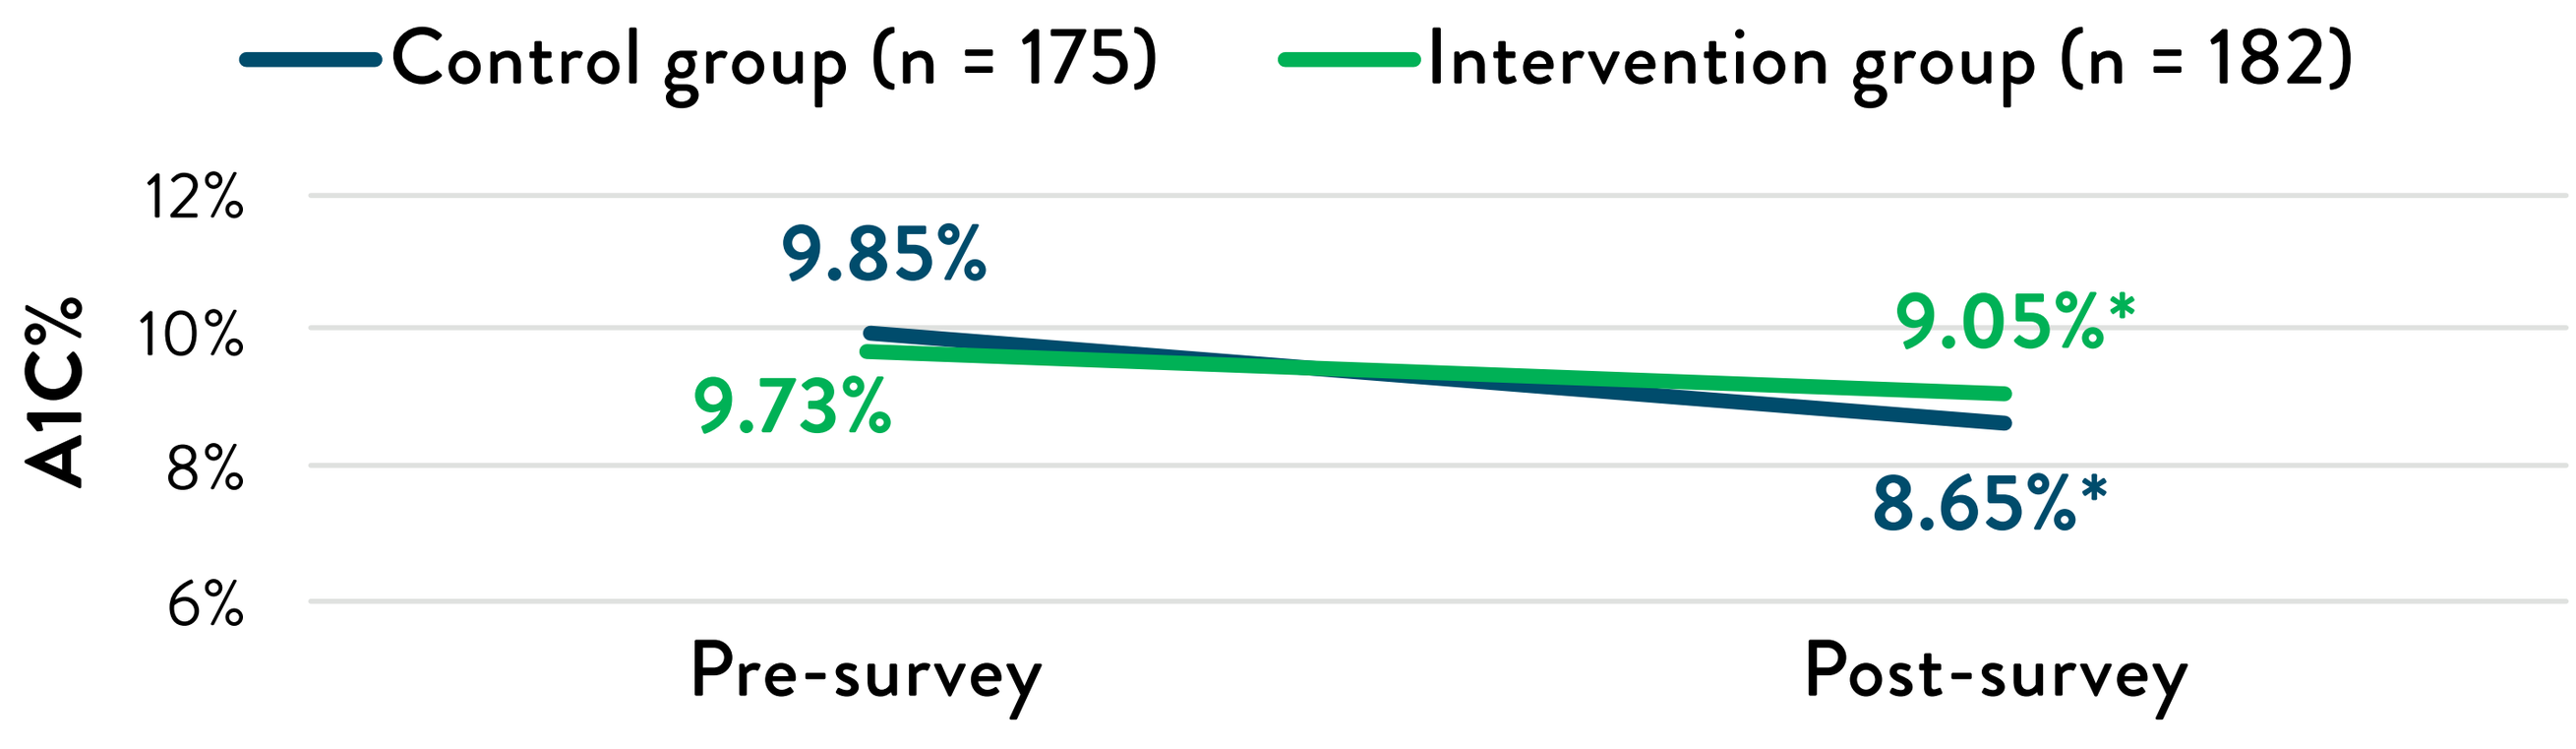
Control group (n = 175)
Intervention group (n = 182)
12%
10%
8%
6%
A1C%
9.85%
9.73%
9.05%*
8.65%*
Pre-survey
Post-survey

*Indicates change from pre- to post-survey was significant based on a paired t-test, p<0.05

Figure 3. Eating Behaviors (Mean Times per Day), by Group, Pre- and Post-survey

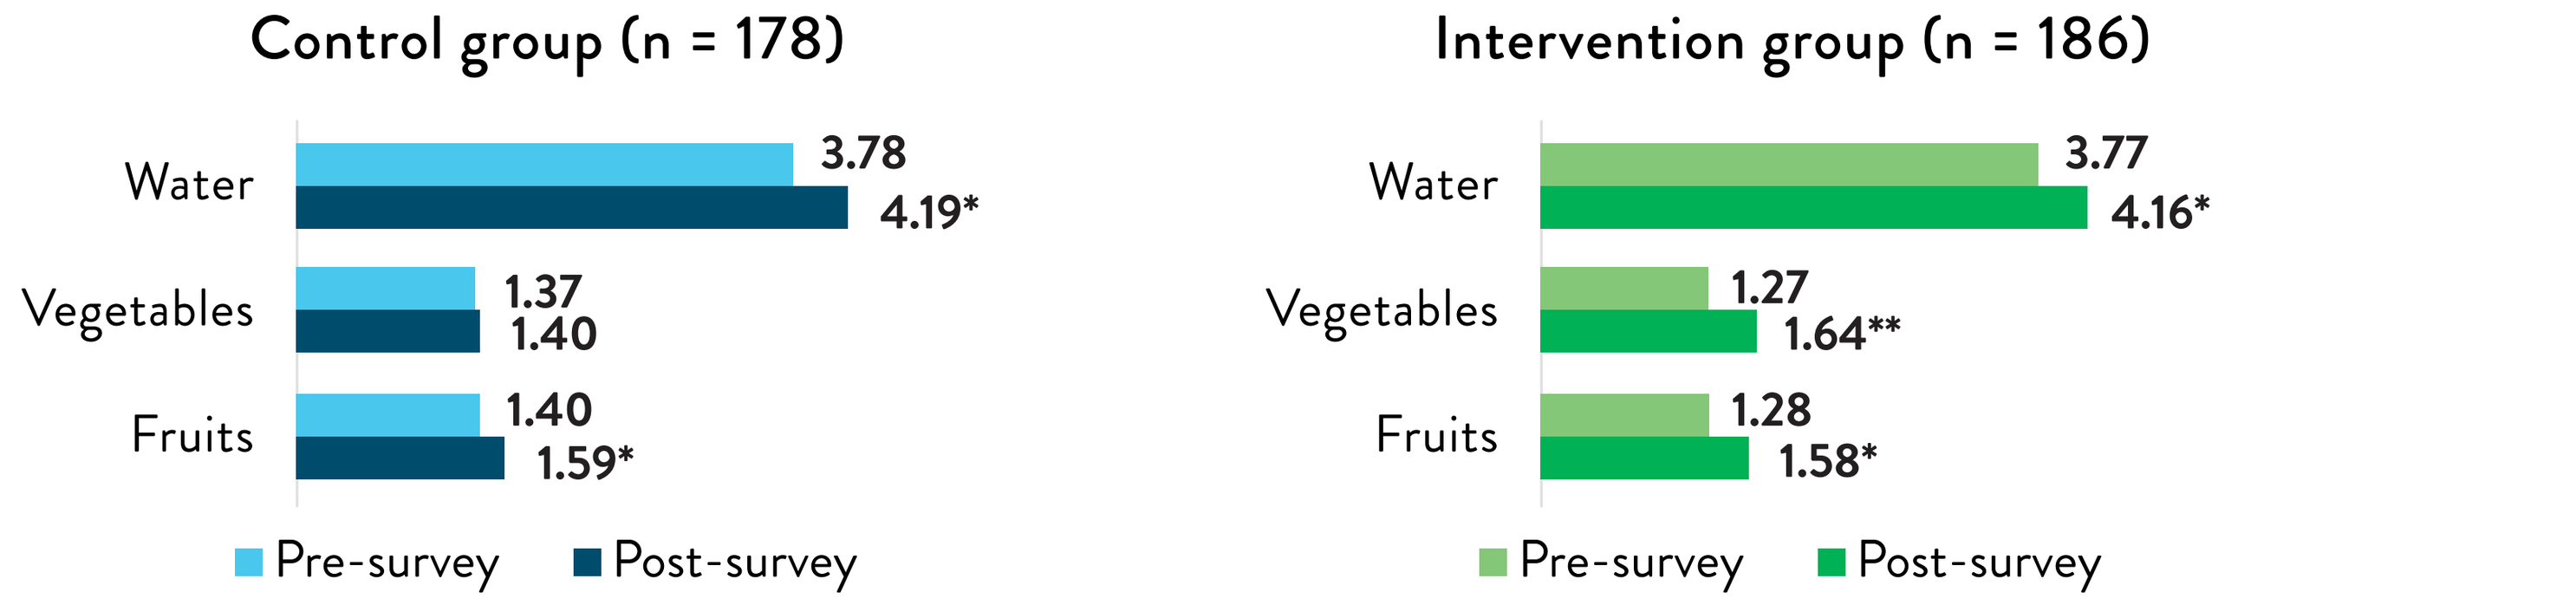
Control group (n = 178)
Water
Vegetables
Fruits
3.78
4.19*
1.37
1.40
1.40
1.59*
Pre-survey
Post-survey
Intervention group (n = 186)
Water
Vegetables
Fruits
3.77
4.16*
1.27
1.64**
1.28
1.58*
Pre-survey
Post-survey

* Indicates change from pre- to post-survey was significant based on a paired t-test, p<0.05
** Indicates change from pre- to post-survey was significant and difference between Intervention and Control group was significant based on a longitudinal mixed model (p=0.007)

Figure 4. Self-reported Health Status (% reporting Good, Very Good, or Excellent), by Group, Pre- and Post-survey

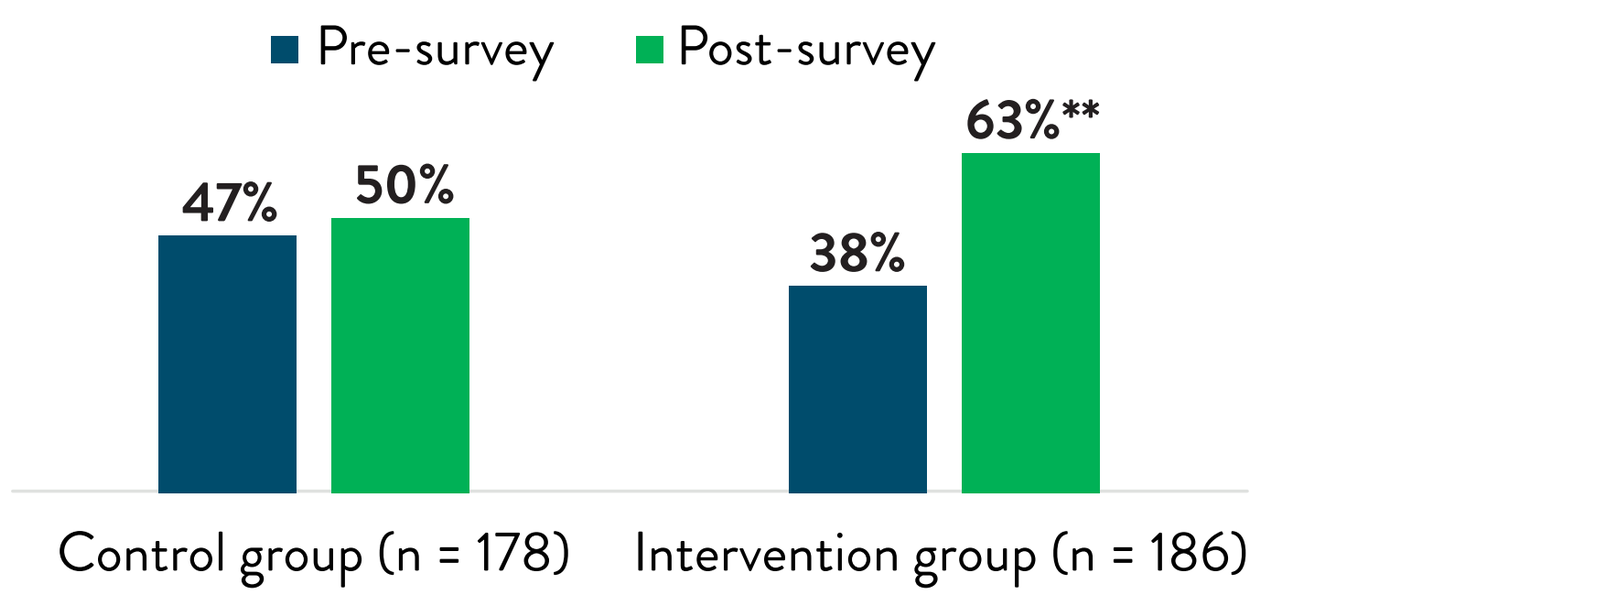
Pre-survey
Post-survey
47%
50%
38%
63%**
Control group (n = 178)
Intervention group (n = 186)

** Indicates change from pre- to post-survey was significant and difference between Intervention and Control group was significant based on a longitudinal mixed model (p<0.001)

Figure 5. Food Security (% of respondents), by Group, Pre- and Post-survey

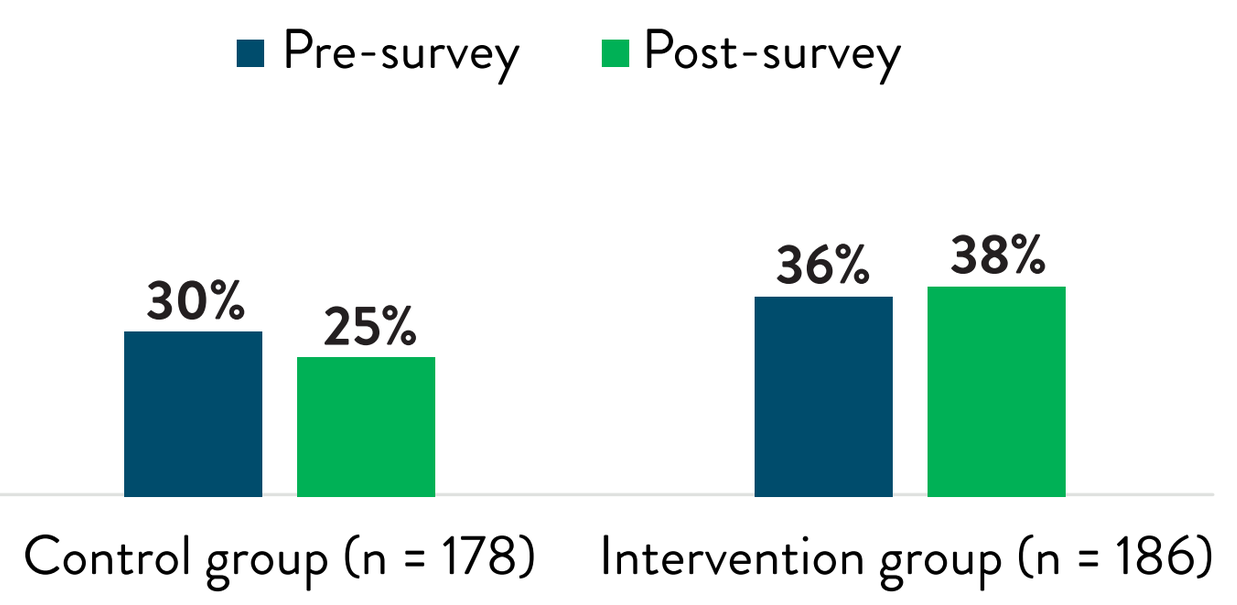
Pre-survey
Post-survey
30%
25%
36%
38%
Control group (n = 178)
Intervention group (n = 186)

CONCLUSIONS

- There were significant improvements in both the Intervention and Control groups, however the Intervention group improved significantly more in vegetable consumption and self-reported health status.
- All participants had access to usual care including access to health educators and other diabetes management support as part of the program which could have contributed to the Control group's positive outcomes.
- Additionally, many participants displayed A1C values out of the target range at baseline, which likely triggered medical interventions, such as medication adjustments to improve A1C values.
- Participant feedback indicates that engaging in the Healthy Food Rx program had a positive impact on diabetes self-management and improved self-efficacy to manage diabetes.

"Everything was perfect. It helped me a lot with my diabetes. Fruits and veggies provided helped me with my sugar levels along with my medications. I feel much better. The food boxes helped me because I did not have a stable job. Thank you very much for the help."
– Healthy Food Rx participant

IMPLICATIONS FOR PRACTICE

- Low-touch Food Is Medicine programs increase access to healthy foods and lead to improvements in some diabetes-related health outcomes that are associated with long-term health benefits.
- While improvements in A1C do not seem to be caused by Food Is Medicine programs alone [4,5], there do seem to be health benefits that could lead to long-term improved chronic disease outcomes. Food Is Medicine programs continue to be a promising health care intervention, although further exploration is needed to understand the best program designs.
- Investing in health educators with dedicated time to support diabetes patients via programs that address social needs can positively impact quality of life and long-term diabetes management.

REFERENCES

1. Centers for Disease Control and Prevention (CDC). About Social Determinants of Health (SDOH) 2021 [Available from: https://www.cdc.gov/socialdeterminants/about.html].
2. San Joaquin County. San Joaquin County 2022 Community Health Needs Assessment. 2022.
3. Gucciardi E, Vahabi M, Norris N, Del Monte JP, Farnum C. The Intersection between Food Insecurity and Diabetes: A Review. Curr Nutr Rep. 2014;3(4):324-32.
4. Doyle J, Alsan M, Skelley N, Lu Y, Cawley J. Effect of an Intensive Food-as-Medicine Program on Health and Health Care Use: A Randomized Clinical Trial. JAMA Intern Med. 2024;184(2):154–163.
5. Shrodes JC, Williams A, Nolan TS, Radabaugh JN, Braun A, Kline D, Zhao S, Brock G, Garner JA, Spees CK, Joseph JJ. Feasibility of Cooking Matters for Diabetes: A 6-week Randomized, Controlled Cooking and Diabetes Self-Management Education Intervention. J Acad Nutr Diet. 2023 Mar;123(3):492-503.e5.

PUBLIC HEALTH INSTITUTE® | CENTER FOR WELLNESS & NUTRITION | Abbott